CHARACTERISATION OF CYTOCHROME P450 INDUCTION IN HEPA RG CELLS

KAJSA P. PERSSON[1,2] AND TOMMY B. ANDERSSON[1,3]

[1]Development DMPK & Bioanalysis, AstraZeneca R&D Mölndal, 431 83 Mölndal, Sweden, [2]Division of Clinical Pharmacology, Department of Laboratory Medicine, Karolinska University Hospital, Huddinge, 141 86 Stockholm, Sweden, [3]Section of Pharmacogenetics,Department of Physiology and Pharmacology, Karolinska Institutet, 171 77 Stockholm, Sweden

Introduction

The cytochrome P450 (P450) enzymes have been shown to be responsible for metabolism of the majority of therapeutics and are therefore important for first pass elimination (bioavailability), clearance and drug-drug interactions. The drug-drug interactions involve inhibition and induction of P450s where enzyme induction can lead to decreased exposure to co-administered drugs, with potential loss of efficacy. Since induction is subjected to important species differences, human hepatocytes has so far been the gold standard model for induction studies. Well known problems with human hepatocytes is the scarce availability, interindividual variability and the need to evaluate multiple donors, which makes the model a non high-throughput model. Because of this, work is ongoing deriving cell lines expressing P450s that could be used for induction studies in early drug discovery.

HepaRG cells is a new human hepatoma cell line that resembles human hepatocytes to a high degree. HepaRG cells express P450s, conjugating enzymes, nuclear receptors and hepatic drug transporters and differentiate into a hepatocyte like morphology (Aninat et al., 2006; Le Vee et al., 2006). They have also been shown to respond to prototypical P450 inducers such as 3-methylcholantrene, rifampicin, and isoniazid (Aninat et al., 2006).

In this study we have investigated the effect of culture time on the mRNA expression of a set of 48 genes in the HepaRG cells. The time course of induction of well-known inducers has also been studied by analysis of mRNA for eight P450s and in addition enzyme activities for three P450s.

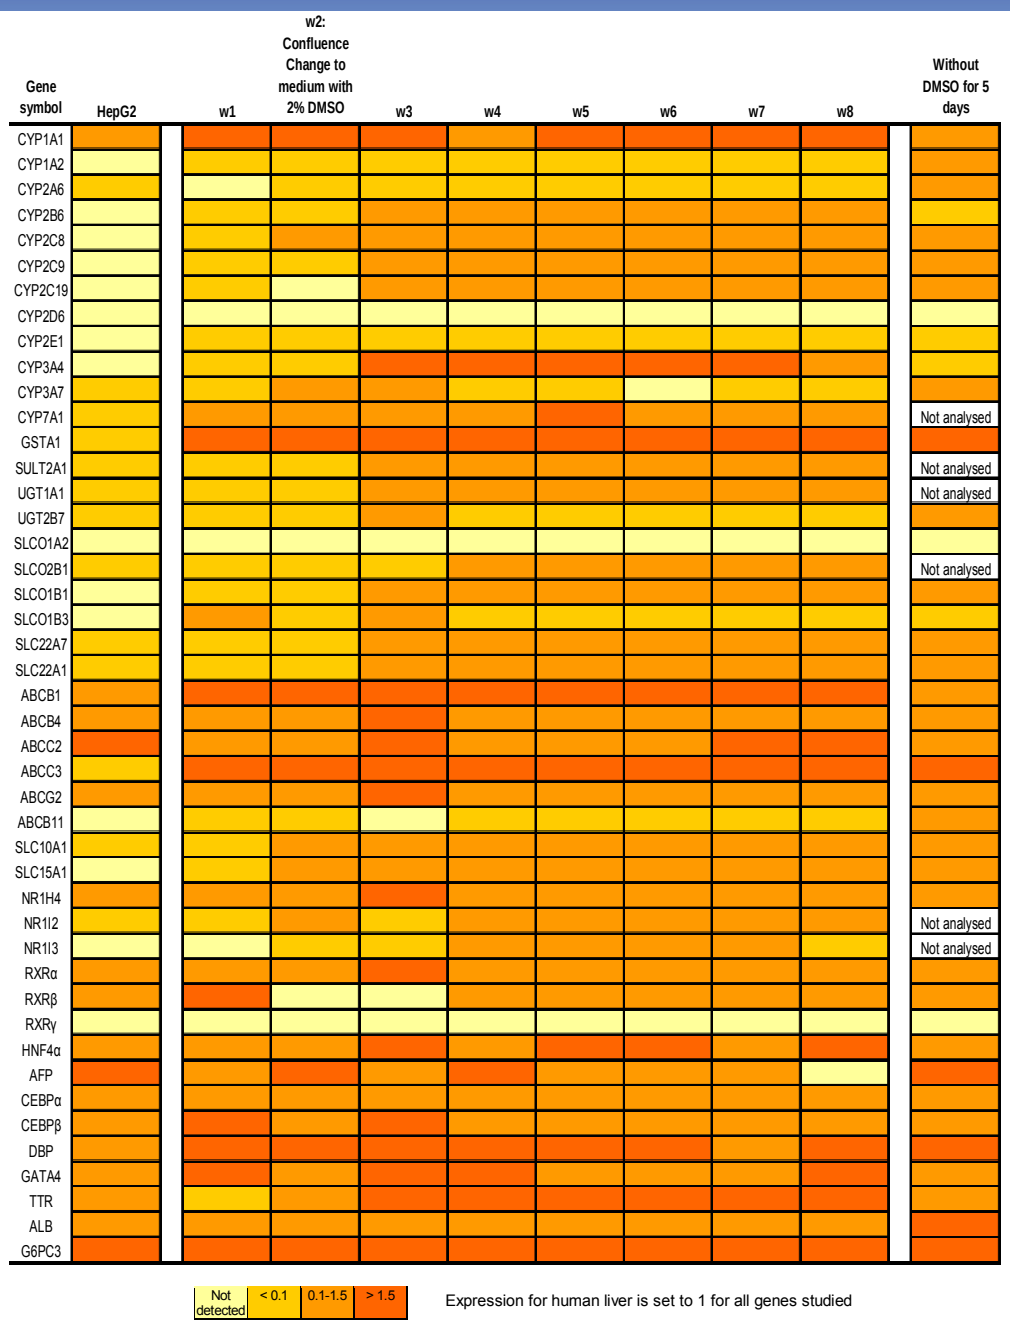
Gene symbol
HepG2
w1
w2: Confluence Change to medium with 2% DMSO
w3
w4
w5
w6
w7
w8
Without DMSO for 5 days
CYP1A1
CYP1A2
CYP2A6
CYP2B6
CYP2C8
CYP2C9
CYP2C19
CYP2D6
CYP2E1
CYP3A4
CYP3A7
CYP7A1
GSTA1
SULT2A1
UGT1A1
UGT2B7
SLCO1A2
SLCO2B1
SLCO1B1
SLCO1B3
SLC22A7
SLC22A1
ABCB1
ABCB4
ABCC2
ABCC3
ABCG2
ABCB11
SLC10A1
SLC15A1
NR1H4
NR1I2
NR1I3
RXRα
RXRβ
RXRγ
HNF4α
AFP
CEBPα
CEBPβ
DBP
GATA4
TTR
ALB
G6PC3
Not analysed
Not detected
< 0.1
0.1-1.5
> 1.5
Expression for human liver is set to 1 for all genes studied

Expression over time of culture

We examined the gene expression in HepaRG cells cultured for 1-8 weeks. Eight of twelve P450s examined were well expressed during week 3 through 8 and the expression of CYP1A1 and 3A4 was considerably higher than for human liver. A relatively low expression was detected for the conjugating enzymes except glutathione S-transferase A1 (GSTA1). The liver membrane transporters were generally well expressed during week 3 through 8, except for OATP-A, which could not be detected in any sample but human liver. A relatively low expression of the nuclear receptors PXR and CAR were seen whereas the expression for FXR, RXRα, RXRβ, and HNF4α were higher or comparable to human liver. Alpha-fetoprotein (AFP) was highly expressed, but the levels decreased over time in culture.

For induction studies HepaRG cells are cultured without DMSO in the medium for five days. The expression levels in these cells were similar or higher than levels in human liver for 32 genes of 39 genes analysed.

The mRNA expression was also measured in HepG2 cells. The only P450s detected in HepG2 cells were low levels of CYP2A6, 3A7 and 7A1 and moderate levels of CYP1A1. Low levels were also detected for the conjugating enzymes. The transport proteins were not detected or expressed in low levels in HepG2 cells except for MRP2. The expression of AFP was extremely high in HepG2 cells, >10 000 times higher than in human liver.

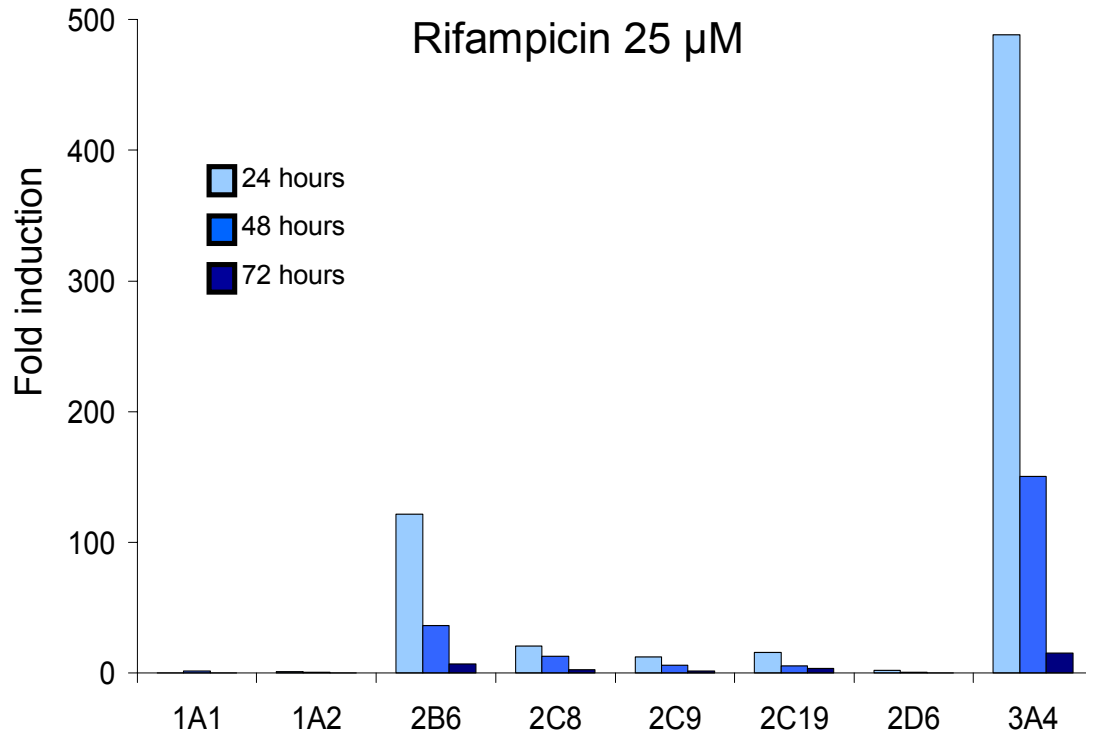
Rifampicin 25 µM
Fold induction
0
100
200
300
400
500
24 hours
48 hours
72 hours
1A1
1A2
2B6
2C8
2C9
2C19
2D6
3A4

Induction of mRNA

HepaRG cells were induced for 24, 48, and 72 hours with rifampicin (25 µM), omeprazole (10 µM), phenobarbital (200 µM), and TCDD (1.55 nM) and mRNA levels were analysed for eight P450s. For all but CYP1A1 the highest expression were seen after 24 hours independent of inducer.

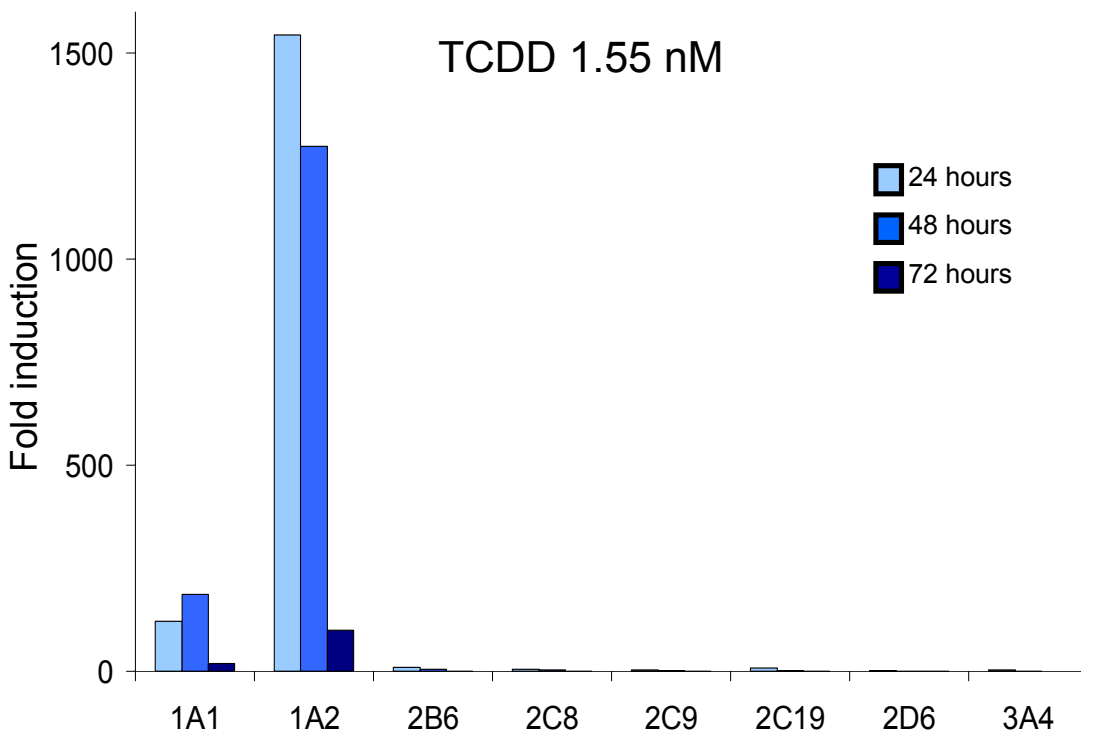
TCDD 1.55 nM
Fold induction
0
500
1000
1500
24 hours
48 hours
72 hours
1A1
1A2
2B6
2C8
2C9
2C19
2D6
3A4

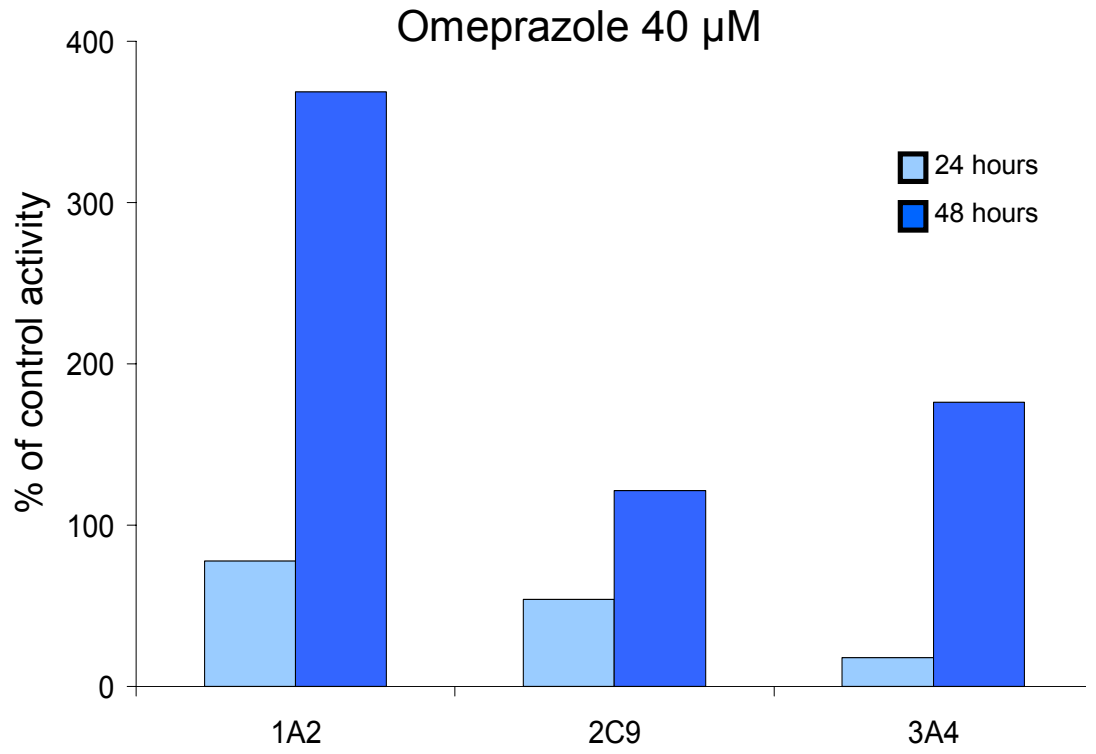
Omeprazole 40 µM
% of control activity
0
100
200
300
400
24 hours
48 hours
1A2
2C9
3A4

Induction of activity

HepaRG cells were induced for 24 and 48 hours with rifampicin (4 µM), omeprazole (40 µM), phenobarbital (200 µM), and primaquine (40 nM) and enzyme activities were analysed for CYP1A2, 2C9, and 3A4. Weak induction of CYP3A4 could be seen already after 24 hours. After 48 hours the induction was evident for all three enzymes.

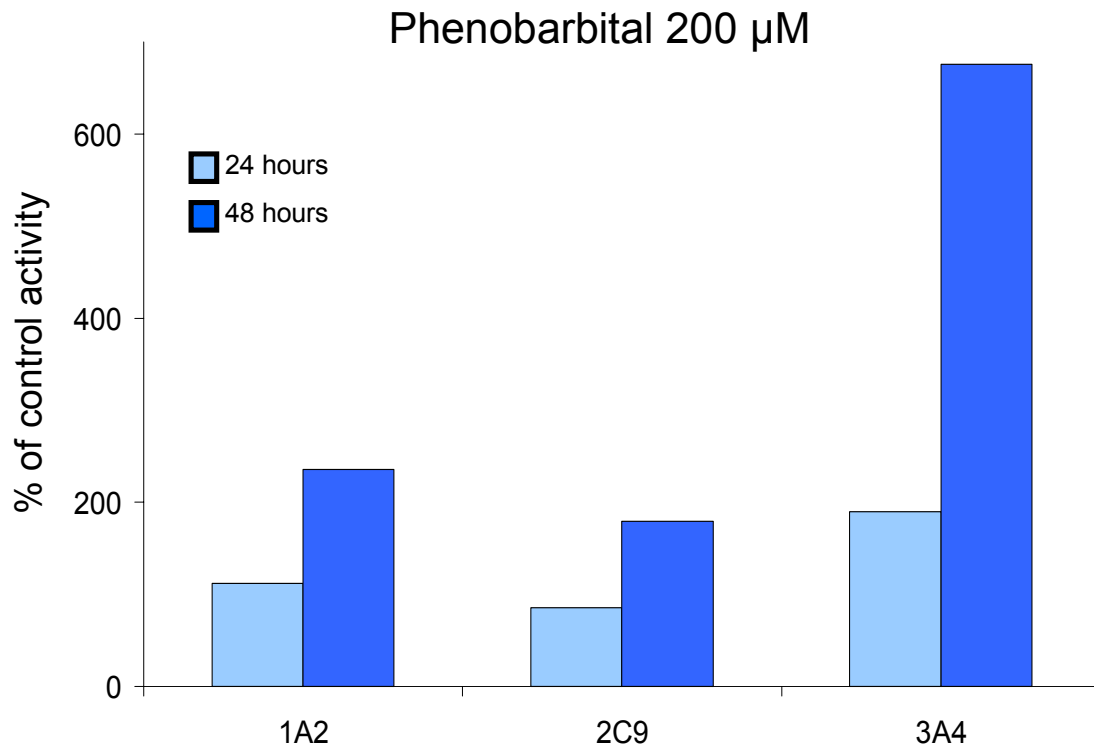
Phenobarbital 200 µM
% of control activity
0
200
400
600
24 hours
48 hours
1A2
2C9
3A4

References

Aninat C, Piton A, Glaise D, Le Charpentier T, Langouet S, Morel F, Guguen-Guillouzo C and Guillouzo A (2006) Expression of cytochromes P450, conjugating enzymes and nuclear receptors in human hepatoma HepaRG cells. *Drug Metabolism & Disposition* 34:75-83.

Le Vee M, Jigorel E, Glaise D, Gripon P, Guguen-Guillouzo C and Fardel O (2006) Functional expression of sinusoidal and canalicular hepatic drug transporters in the differentiated human hepatoma HepaRG cell line. *European Journal of Pharmaceutical Sciences* 28:109-117.

Conclusion

The stable expression of mRNA in HepaRG cells between week 3 through 8 makes it a robust cell line. In combination with the expression of P450s, transport proteins and liver specific markers this makes the HepaRG cells useful for experimentation.

HepaRG cells are induced by many prototypical P450 inducers, which makes it suitable for induction studies.